SAN ANTONIO BREAST CANCER SYMPOSIUM – CANCER THERAPY AND RESEARCH CENTER AT UT HEALTH SCIENCE CENTER – DECEMBER 10-14, 2013

Neoadjuvant chemotherapy in gynecologic oncology practices in Germany: a real life documentation

Joerg Schilling[1], Hans Joachim Hindenburg[2] for the German Professional Association of Gynecologic Oncology in Practices (BNGO)

1 BNGO e.V., Neuenhagen b. Berlin; 2 Practice for Gyneco-Oncology, Berlin

Background

In patients with advanced, primarily inoperable breast cancer neoadjuvant chemotherapy (NACT) can achieve operability and potentially reduce the radicalism of the operation. Additionally, several clinical studies have demonstrated that NACT is equally effective to adjuvant chemotherapy in patients with early breast cancer. The advantages of NACT include potentially improved rates of breast conserving surgery, the possibility of measuring early, in-vivo response to systemic treatment, and potentially improved outcomes for certain subgroups of high-risk breast cancer patients.

In Germany, many patients with early breast cancer receive systemic therapy in an outpatient setting. The German Professional Association of Gynecologic Oncology in Practices (BNGO) is dedicated to quality-assured outpatient treatment of patients with gynecologic tumors. This includes adjuvant and neoadjuvant chemotherapy.

Since 1/1/2004, BNGO member practices have documented their patients in a specialized computerized documentation system (ODM QuaSi®GYN). All practices participate in this registry on a voluntary basis. The purpose of the documentation is to gain and compare patient details and treatment behaviors in the BNGO group. A second objective is the voluntary documentation of treatment quality within the BNGO practices, which will become mandatory by the health care authorities in Germany in the near future.

Objectives

In this retrospective study we analyzed the details of breast cancer patients who were treated with neoadjuvant chemotherapy (plus/minus anti-HER2-therapy or other targeted therapy) in 102 BNGO practices between 2004 and 2013. Our objectives were to evaluate the patient characteristics of the breast cancer patients treated with NACT in our practices, the most frequently applied regimens for NACT, the clinical efficacy and toxicity of these regimens and the rate of breast conserving operation after NACT and the event free survival rates. Pathological responses after surgery were not analyzed, because operations were not performed in the BNGO practices and therefore were not assessed in this system.

Methods

From January 2004 to November 2013, a total of 21,598 breast cancer patients in 102 practices were recorded in the BNGO data base registry. For documentation, the specialized ODM QuaSi®GYN online documentation system is used. The documentation includes demographic data of individual patients and practices as well as data regarding diagnosis, treatment and outcome.

In this analysis we present the data of 2,603 breast cancer patients (12 %) who received neoadjuvant chemotherapy in BNGO practices.

Patient characteristics (n = 2,603 patients)

Age

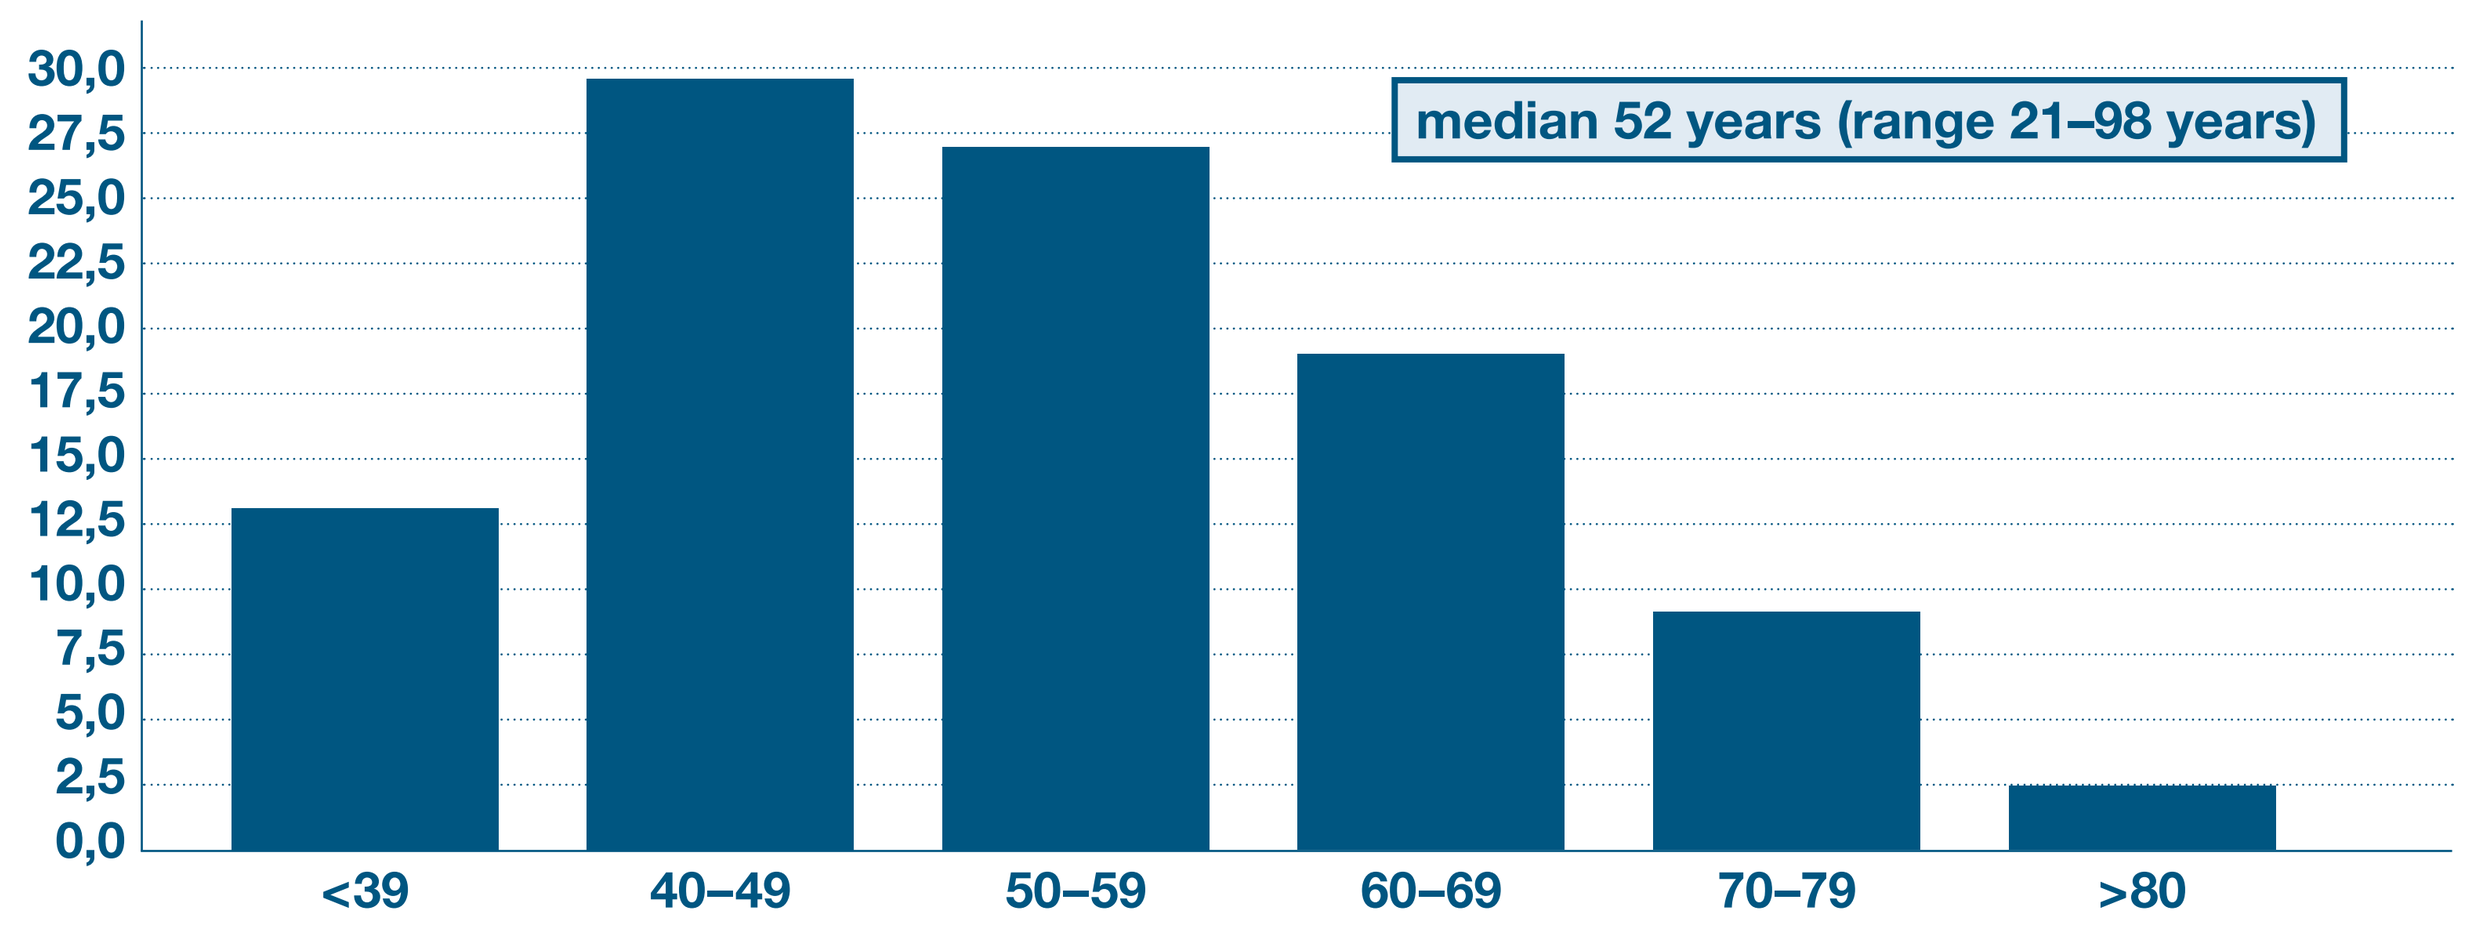
median 52 years (range 21–98 years)
30,0
27,5
25,0
22,5
20,0
17,5
15,0
12,5
10,0
7,5
5,0
2,5
0,0
<39
40–49
50–59
60–69
70–79
>80

Tumor stage (T)

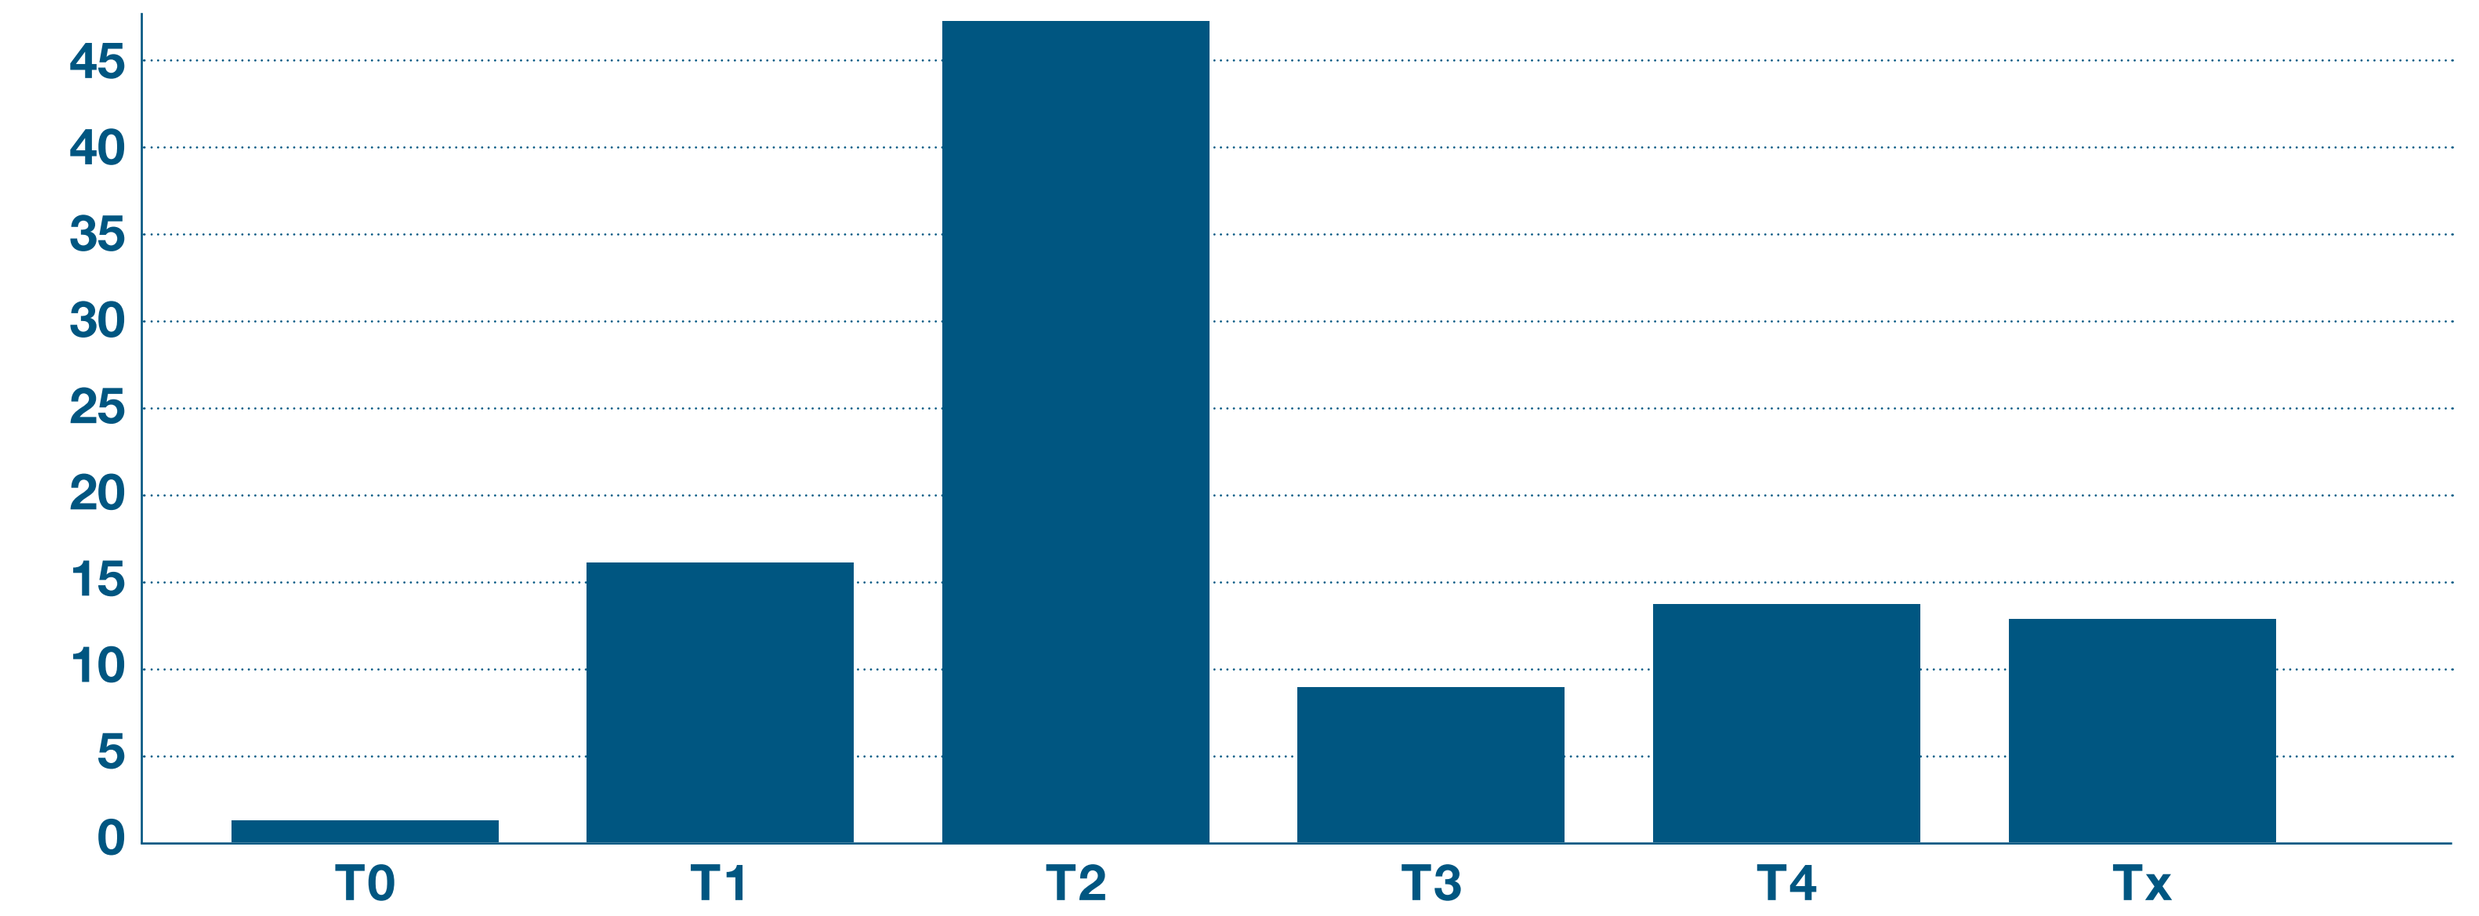
45
40
35
30
25
20
15
10
5
0
T0
T1
T2
T3
T4
Tx

Regional lymph nodes (N)

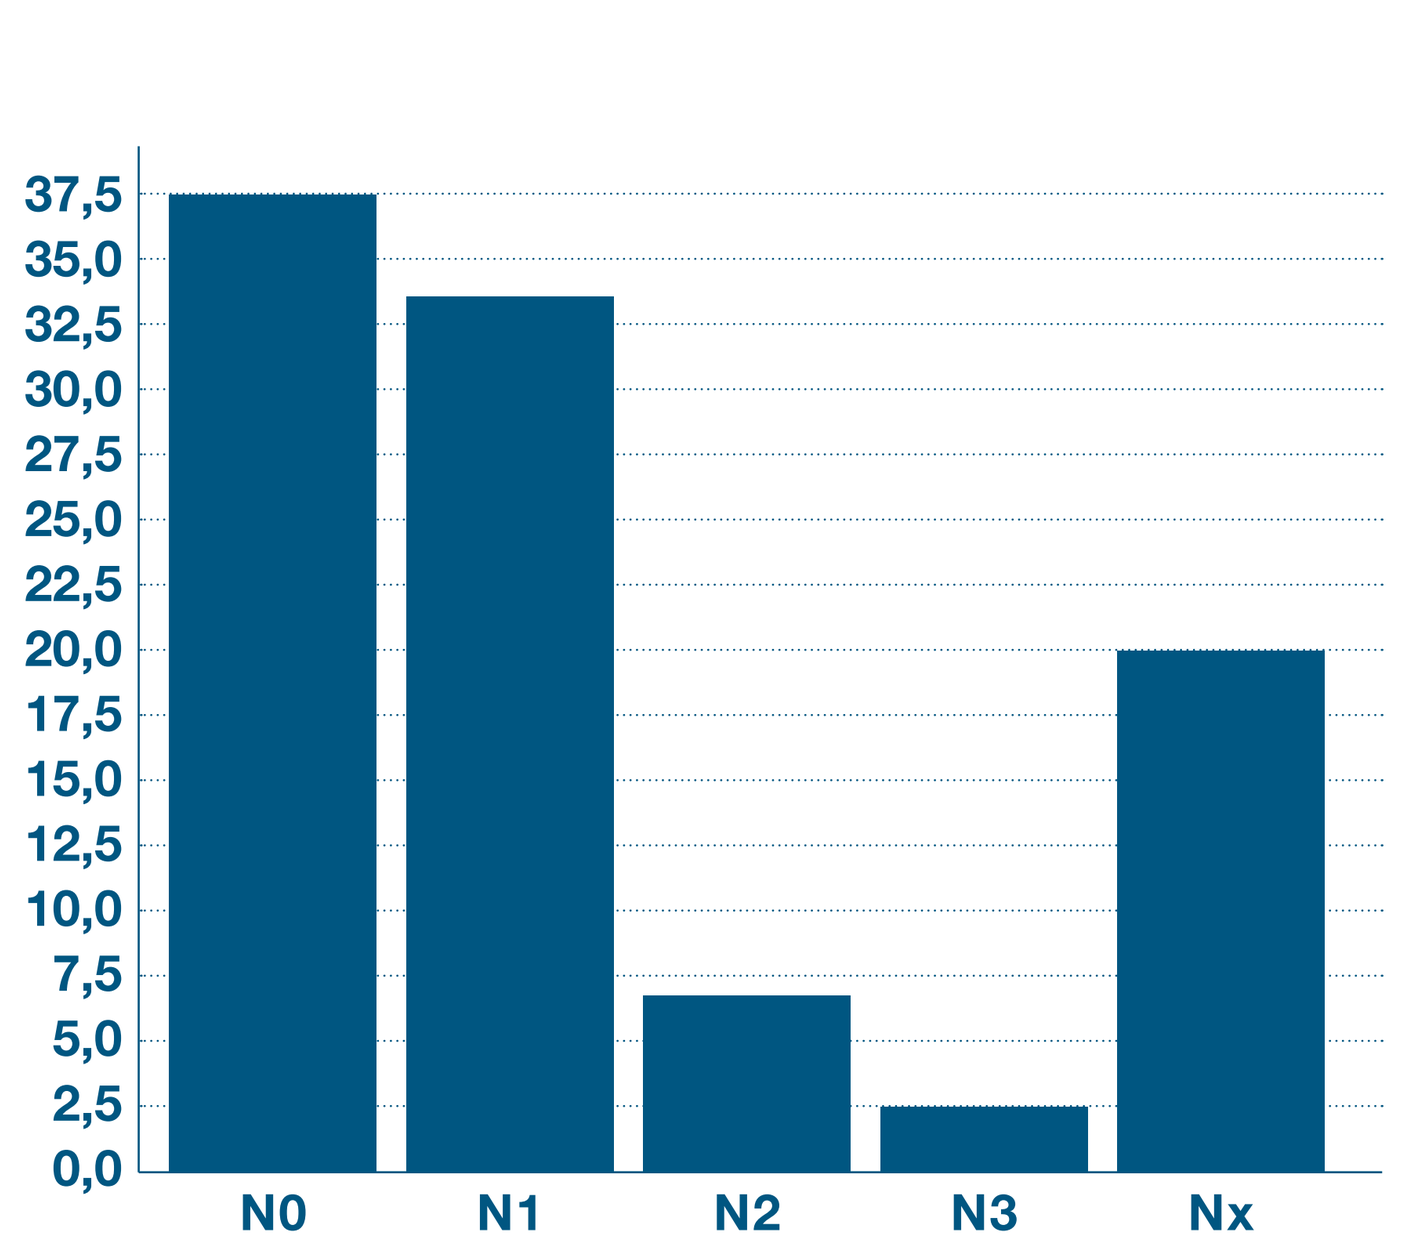
37,5
35,0
32,5
30,0
27,5
25,0
22,5
20,0
17,5
15,0
12,5
10,0
7,5
5,0
2,5
0,0
N0
N1
N2
N3
Nx

Metastases (M)

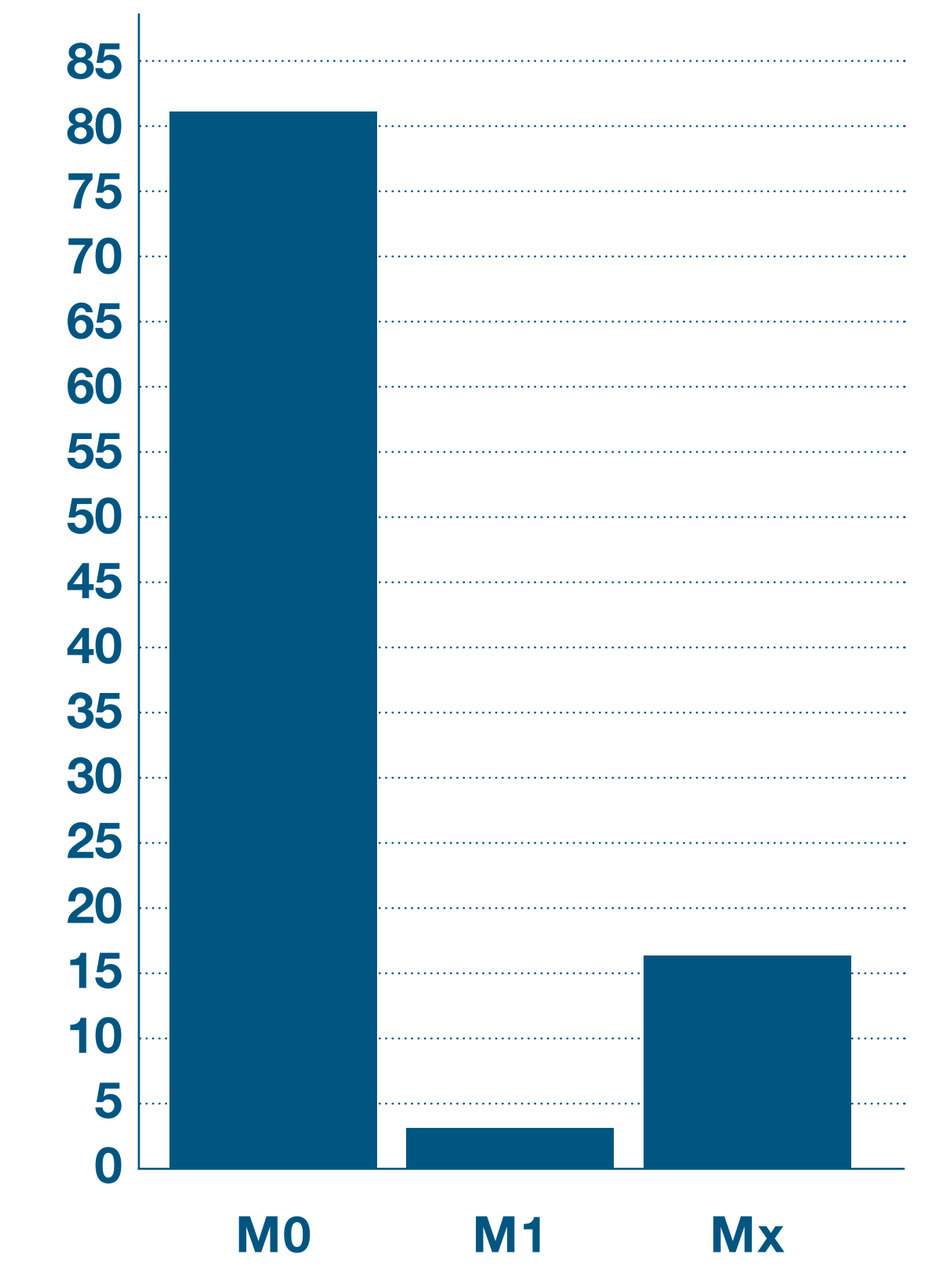
85
80
75
70
65
60
55
50
45
40
35
30
25
20
15
10
5
0
M0
M1
Mx

Hormone receptor- and HER2-status at primary diagnosis of breast cancer

	Number of patients	Percentage
Hormone receptor status		
ER-positive	1,295	50 %
ER-negative	852	33 %
Unknown	456	18 %
PR-positive	1,170	44 %
PR-negative	976	38 %
Unknown	457	18 %
HER2 receptor status		
HER2-positive (IHC 3+)	652	25 %
HER2-negative (IHC 0, 1+, 2+)	1,782	69 %
Unknown	169	6 %

Neoadjuvant chemotherapy:
Treatment regimens – anthracyclines/taxanes (n = 2,603 pts)

Anthracycline-containing regimen	84 %
Taxane-containing regimen	83 %

Most frequently used chemotherapy regimens

Regimen	HER2-positive pts (n = 652)	HER- negative pts (n = 1,782)
EC-Docetaxel (q3w)	113	320
EC-Paclitaxel (weekly)	71	210
TC	64	237
EC	58	123
Pacitaxel weekly	38	85
Docetaxel, Carboplatin	36	Below 10 pts.
EC-Paclitaxel (q3w)	33	Below 10 pts.
TAC	30	165
FEC-Doc (q3w)	21	36
Docetaxel (q3w)	18	33
Paclitaxel (q3w)	18	27
MC	15	40
FEC	14	71
AC-Docetaxel (q3w)	Below 10 pts.	13
Others*	331	677

* All regimens with less than 10 documented pts per protocol are summarized in "others"

A = adriamycin (doxorubicin), C = cyclophosphamide, E = epirubicin, F = 5-fluorouracil, M = mitoxantrone, T = docetaxel

Additional targeted therapy

Additional anti HER2-therapy in HER2-positive patients (n = 652 pts)

471 pts (72 %) received additional trastuzumab
35 pts (5 %) received additional lapatinib

Additional targeted therapy in HER2-negative patients (n = 1782)

88 pts (5 %) received additional bevacizumab

Results

Clinical efficacy
Overall response rate to NACT (2,603 pts)

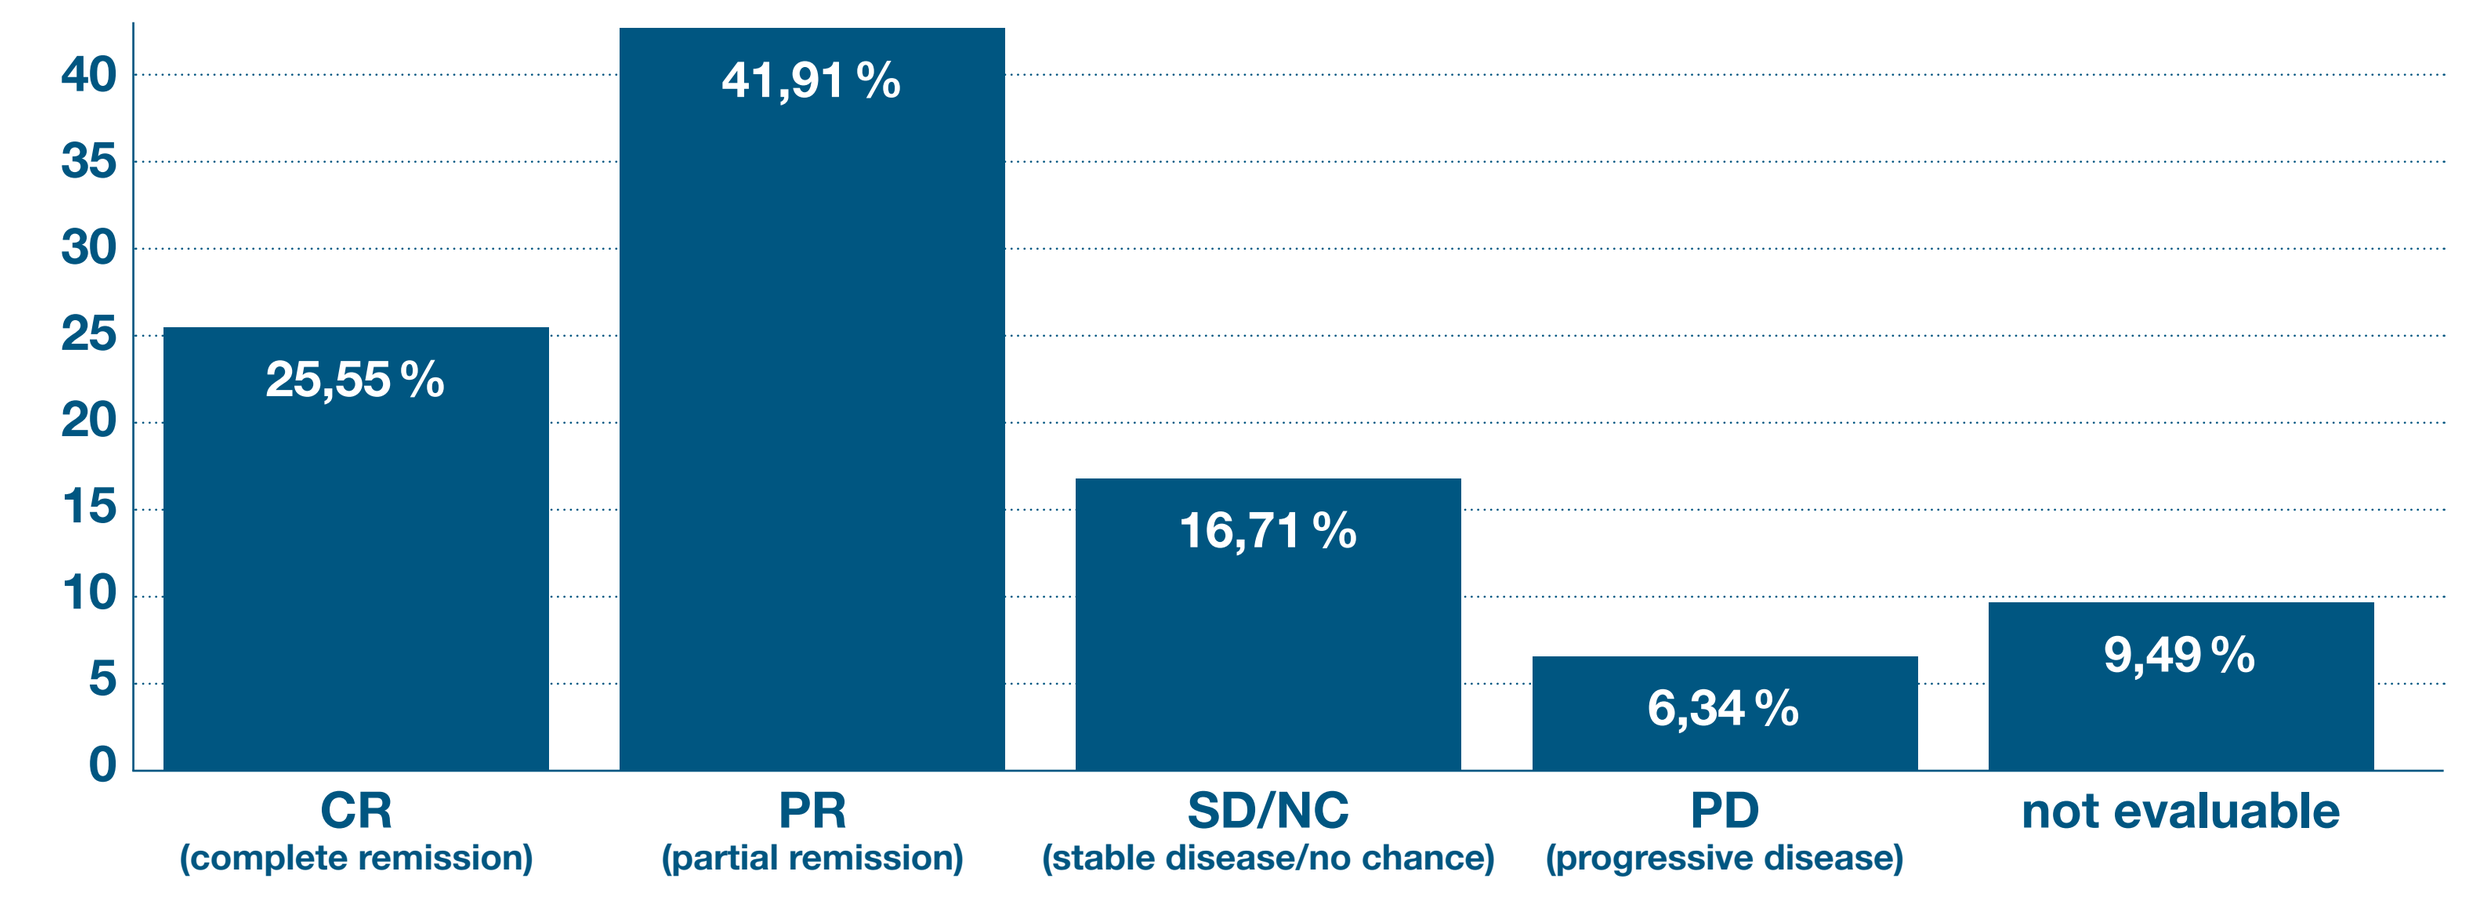
40
35
30
25
20
15
10
5
0
25,55 %
41,91 %
16,71 %
6,34 %
9,49 %
CR
(complete remission)
PR
(partial remission)
SD/NC
(stable disease/no chance)
PD
(progressive disease)
not evaluable

Clinical response to NACT in HER2-negative patients

Regimen	n pts.	CR (%)	PR (%)	SD (%)	PD (%)	n.e. (%)
EC-Doc (q3w)	320	12,19 %	57,50 %	15,63 %	8,44 %	6,25 %
TC	237	26,16 %	43,88 %	19,41 %	5,49 %	5,06 %
EC- Pacl. weekly	210	10,95 %	62,86 %	10,00 %	6,67 %	9,52 %
TAC	165	21,21 %	41,21 %	23,64 %	3,03 %	10,91 %
EC	123	9,76 %	49,59 %	21,14 %	12,20 %	7,32 %
Pacl. weekly	85	15,29 %	41,18 %	17,65 %	10,59 %	15,29 %
FEC	71	19,72 %	54,93 %	12,68 %	5,63 %	7,04 %
EC-Pacl. (q3w)	57	14,04 %	68,42 %	8,77 %	5,26 %	3,51 %
MC	40	27,50 %	32,50 %	15,00 %	10,00 %	15,00 %
FEC-Doc (q3w)	36	36,11 %	33,33 %	19,44 %	2,78 %	8,33 %
Docetaxel (q3w)	33	12,12 %	54,55 %	15,15 %	6,06 %	12,12 %
Paclitaxel (q3w)	27	11,11 %	62,96 %	18,52 %	3,70 %	3,70 %
AC-Doc (q3w)	13	15,38 %	38,46 %	15,38 %	7,69 %	23,08 %

Clinical response to NACT in HER2-positive patients

Regimen	n pts.	CR (%)	PR (%)	SD (%)	PD (%)	n.e. (%)
EC-Docetaxel (q3w)	113	18,58 %	18,58 %	20,35 %	20,35 %	20,35 %
EC-Paclitaxel (weekly)	71	15,49 %	15,49 %	5,63 %	5,63 %	5,63 %
TC	64	34,38 %	39,06 %	17,19 %	4,69 %	4,69 %
EC	58	13,79 %	13,79 %	17,24 %	17,24 %	17,24 %
Pacitaxel weekly	38	15,79 %	15,79 %	7,89 %	7,89 %	7,89 %
Docetaxel, Carboplatin	36	22,22 %	22,22 %	11,11 %	11,11 %	11,11 %
EC-Paclitaxel (q3w)	33	18,18 %	18,18 %	15,15 %	15,15 %	15,15 %
TAC	30	20,00 %	20,00 %	20,00 %	20,00 %	20,00 %
FEC-Doc (q3w)	21	57,14 %	57,14 %	19,05 %	19,05 %	19,05 %
Docetaxel (q3w)	18	27,78 %	27,78 %	27,78 %	27,78 %	27,78 %
Paclitaxel (q3w)	18	22,22 %	22,22 %	27,78 %	27,78 %	27,78 %
MC	15	26,67 %	26,67 %	26,67 %	26,67 %	26,67 %
FEC	14	35,71 %	35,71 %	14,29 %	14,29 %	14,29 %

Breast conserving surgery

78 % breast-conserving therapy and 22 % mastectomy, n = 2603

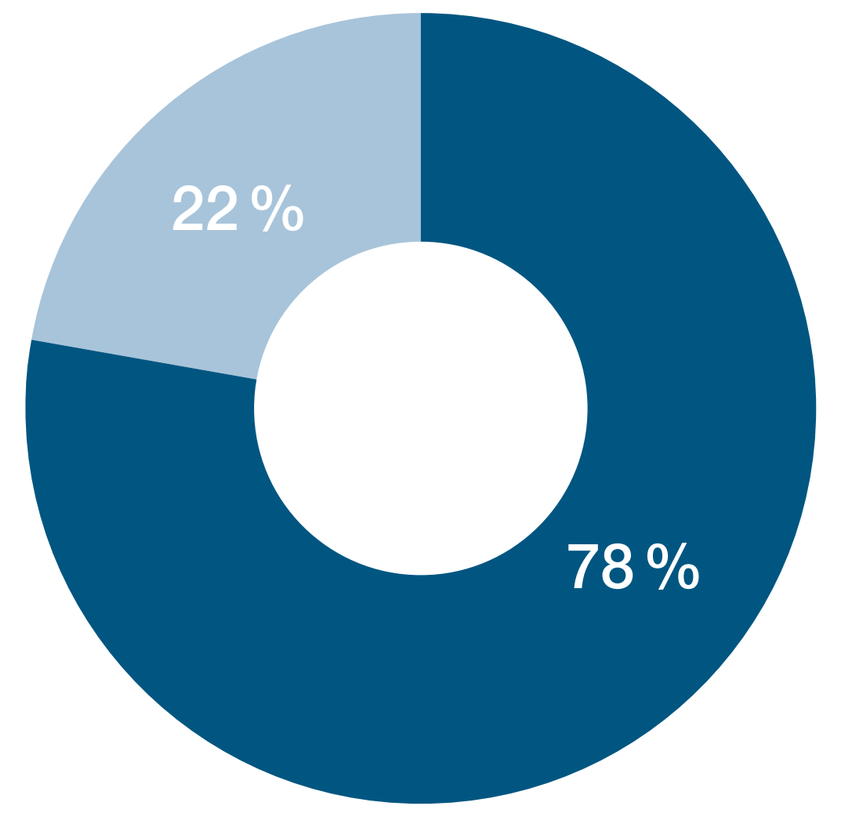
22 %
78 %

Survival

Progression free survival and survival without metastatic disease

All patients since 2004 who were evaluable at the respective time point have been included in the analyis.

Documentation time	Lost to follow up (n pts)	Evaluable (n pts)	% pts without progression or metastases
2 years	0	1591	85
5 years	79	684	75
10 years	125	40	75

Toxicity

A comprehensive supportive care program according to guidelines is applied in the BNGO practices to prevent side effects like neutropenia, anemia or nausea and vomiting. Therefore toxicities of grade III or IV (NCI CTC common toxicity criteria) were rare. In general, severe toxicities (grade III or IV) were observed in less than 15 % of cycles. Dose reductions and cycle delays were required in only 10–15 % of cycles, mostly related to anthracycline toxicity.

Grade III/ IV hematologic toxicities (n = 2,603 pts)

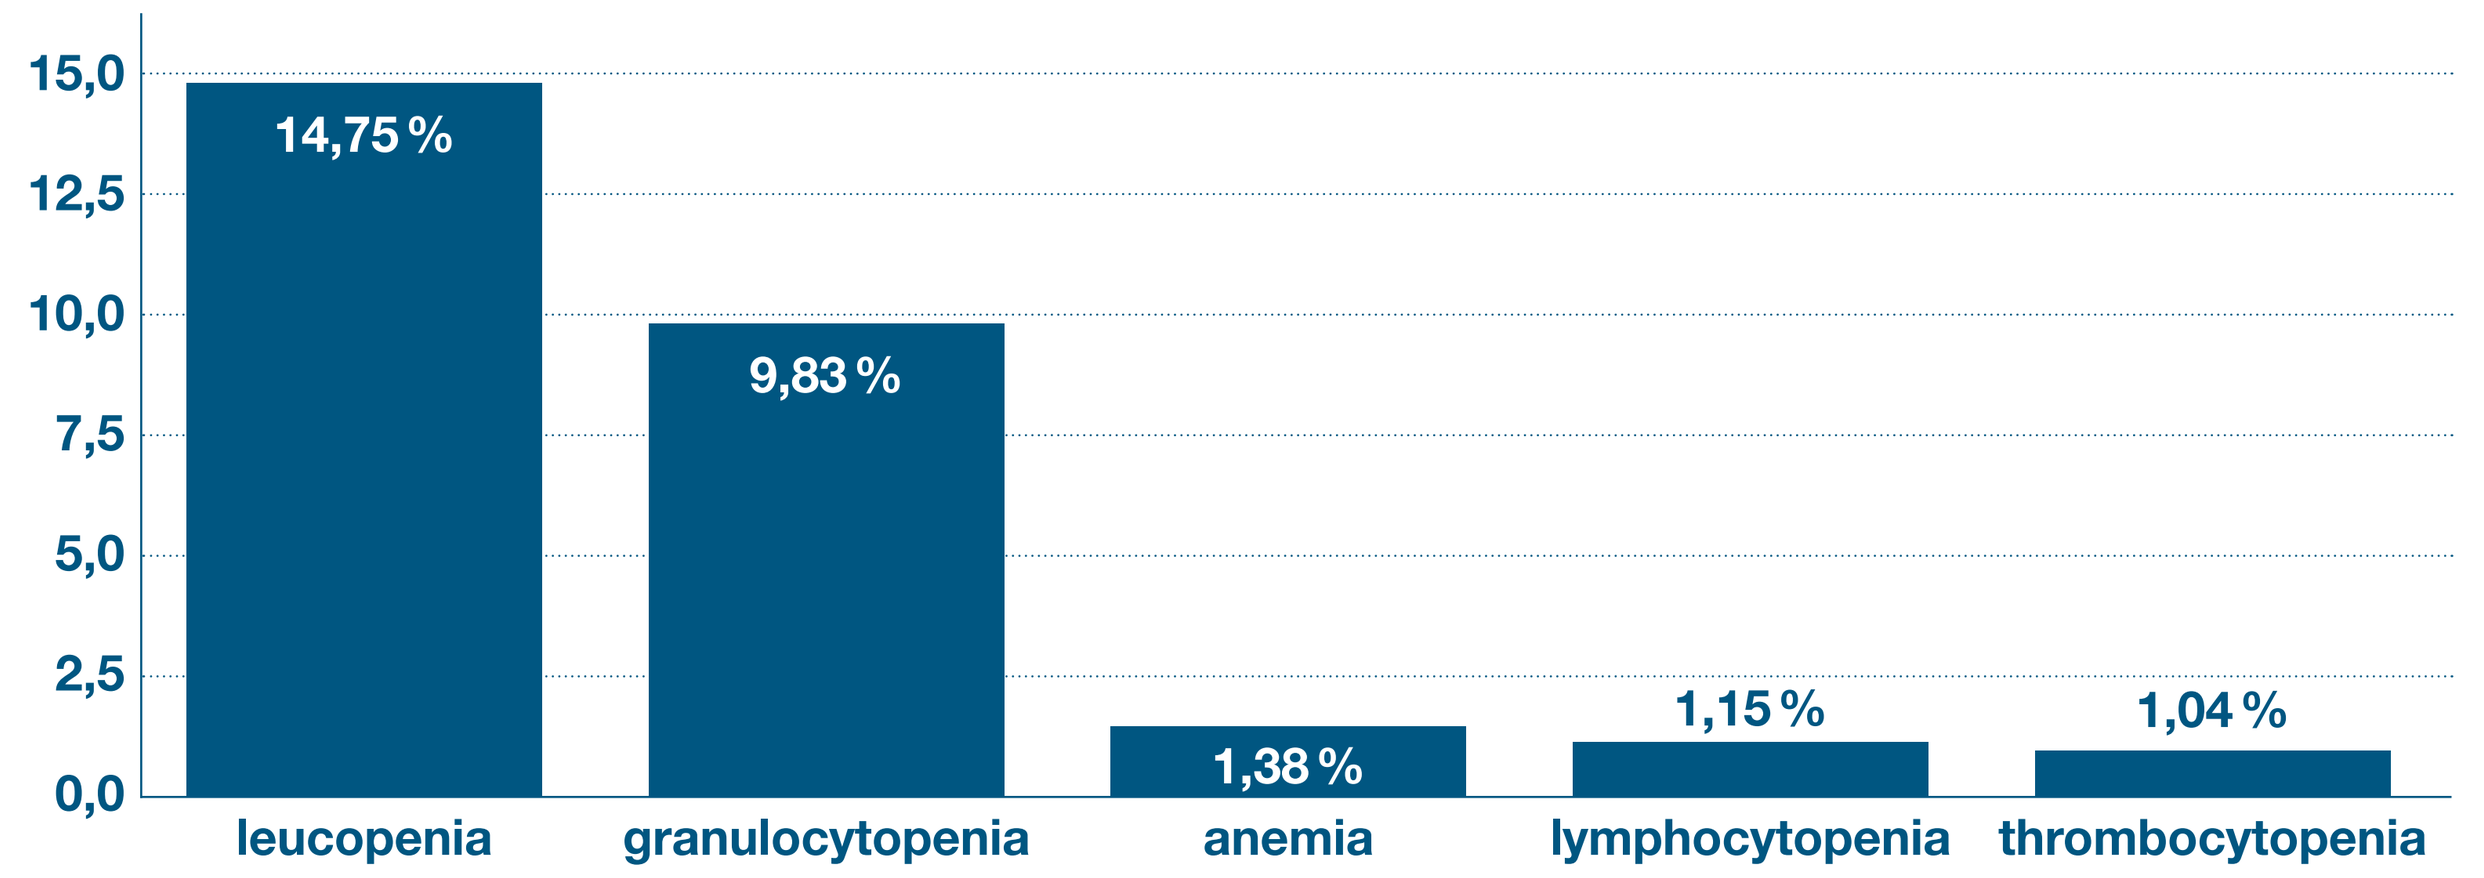
15,0
12,5
10,0
7,5
5,0
2,5
0,0
14,75 %
9,83 %
1,38 %
1,15 %
1,04 %
leucopenia
granulocytopenia
anemia
lymphocytopenia
thrombocytopenia

Grade III/ IV non hematologic toxicities (2,603 pts)

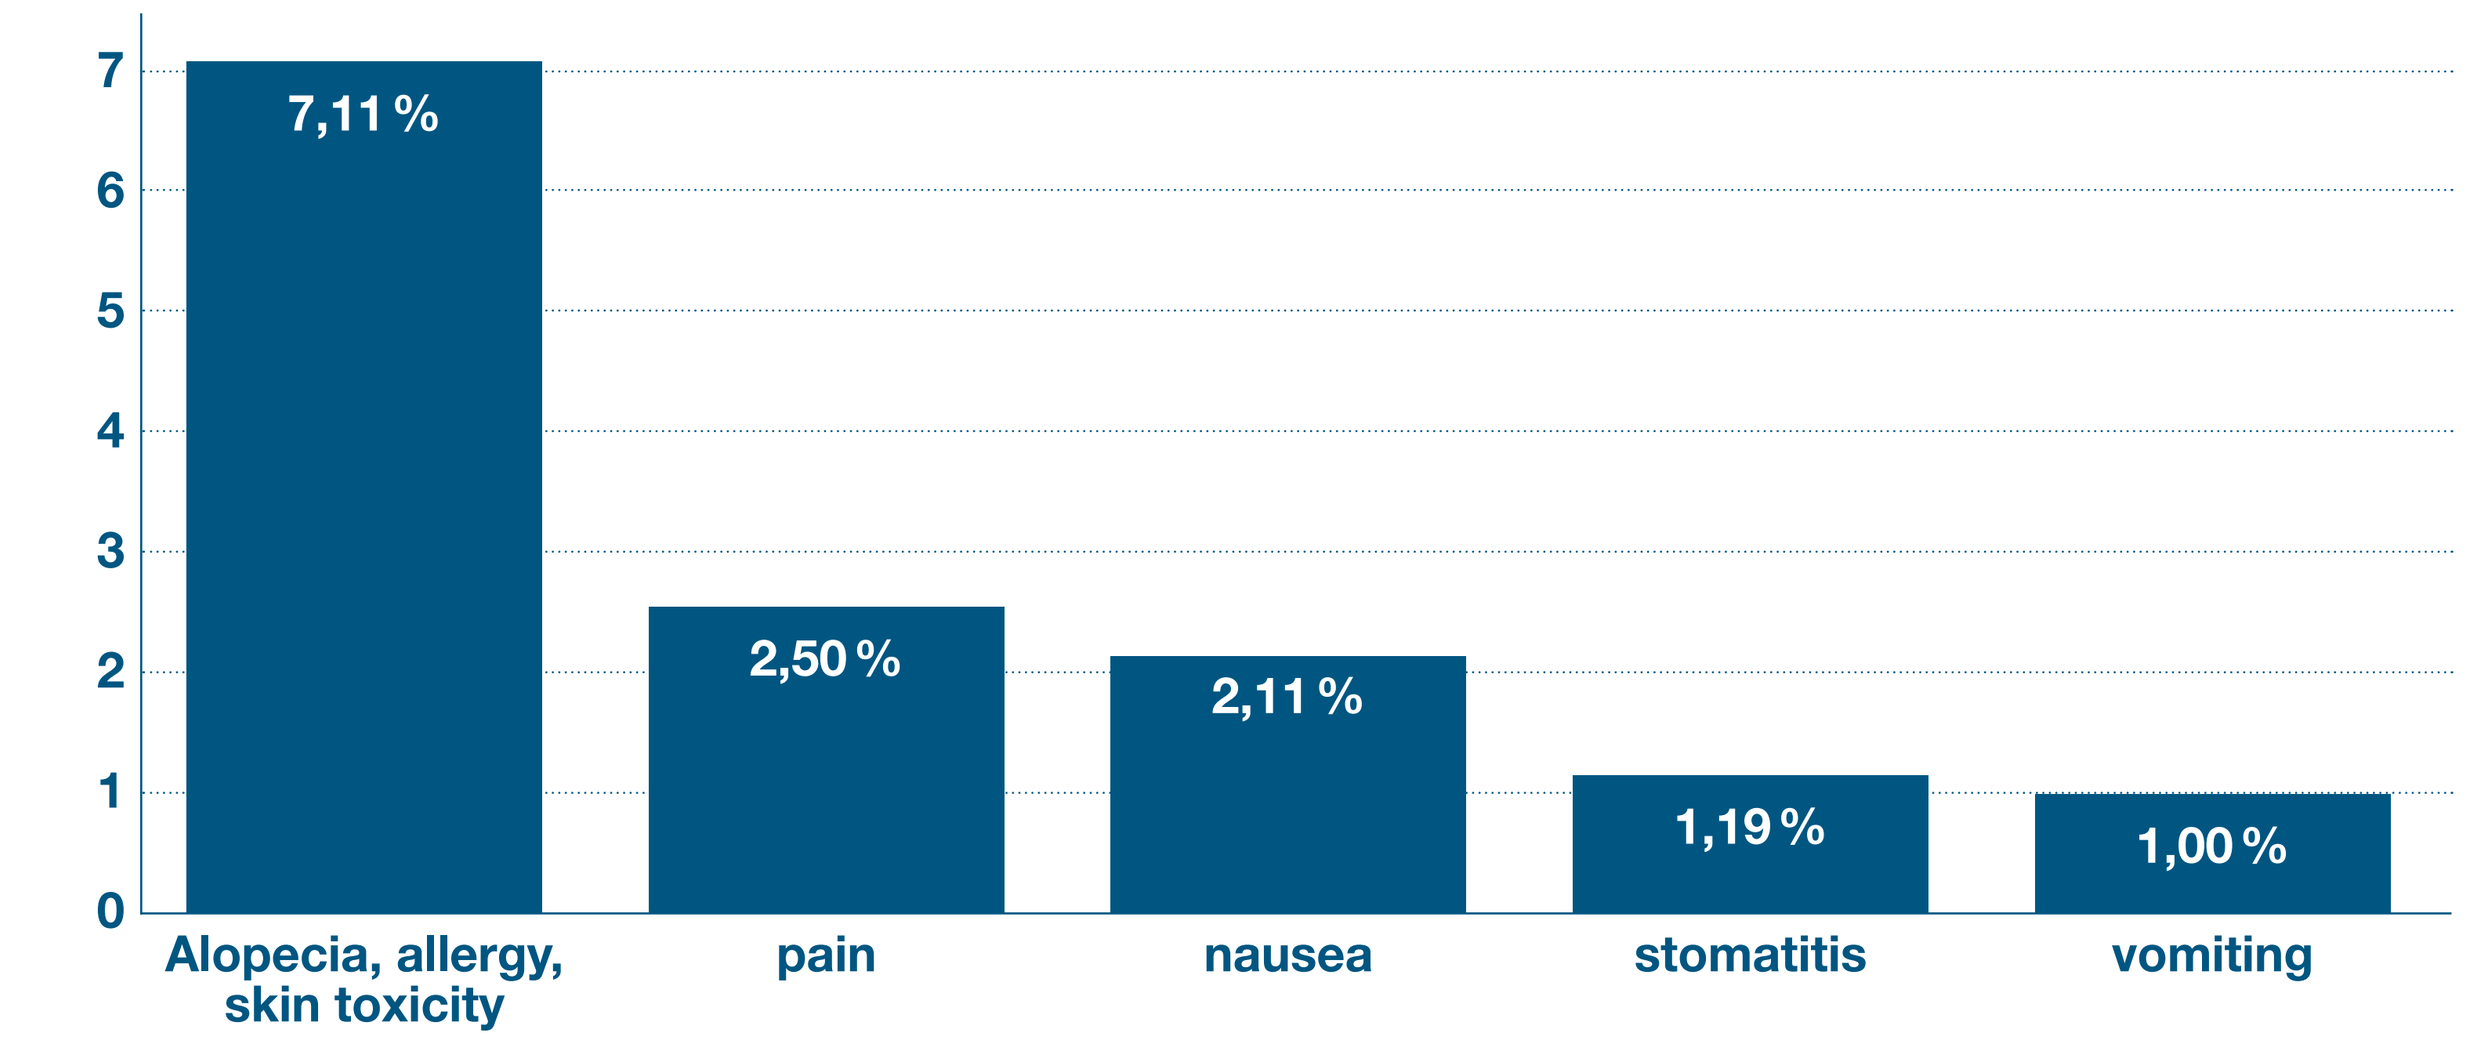
7
6
5
4
3
2
1
0
7,11 %
2,50 %
2,11 %
1,19 %
1,00 %
Alopecia, allergy, skin toxicity
pain
nausea
stomatitis
vomiting

Conclusion

Between January 2004 and November 2013, 2,603 breast cancer patients who received neoadjuvant chemotherapy +/– targeted therapy for breast cancer were treated in BNGO-practices. Most treatment schedules contained an anthracycline and/or a taxane. HER2-positive patients received additionalanti-HER2-therapy. The objective clinical response rate in all patients (PR and CR) was 67 %. After NACT, breast conserving surgery was possible in nearly 80 % of patients. Due to prophylactic supportive care the toxicity was mild with grade III/IV toxicities observed in less than 12 % of patients. The clinical results of this real life documentation with non-selected breast cancer patients compare to results achieved in trials under research conditions with a selected patient population. Breast cancer patients who are treated with neoadjuvant systemic therapy in German BNGO practices can be sure to receive a high quality systemic treatment with a clinical response rate comparable to the results in clinical trials and they have an 80% chance of breast conserving therapy after NACT.

This presentation is the intellectual property of the author/presenter. Contact at info@bngo.de for permission to reprint and/or distribute.

P3-14-21